Exposure-Response Analyses of Efficacy and Safety of Patritumab Deruxtecan in Cancer Patients

PMX618

Rujuta Joshi[1], Shelly Wang[2], Rena Byrne[2], David Sternberg[1], Pomy Shrestha[1], Mark Lee[1], Malaz Abutarif[1], Li Li[1], Tushar Garimella[1]

1. Daiichi Sankyo, Inc., Basking Ridge, NJ, USA. 2. Metrum Research Group, Tariffville, CT, USA

OBJECTIVES

1. To characterize the HER3-DXd exposure-response (ER) relationships between the anti-HER3 antibody conjugated payload (anti-HER3-ac-DXd) and payload (DXd) exposures and efficacy as measured by the Objective response rate (ORR) in NSCLC patients
2. To characterize the ER relationships between anti-HER3-ac-DXd and DXd exposures and safety in patients with NSCLC or BC:
 a) Grade 3+ platelet count decrease based on lab values (TCP)
 b) Grade 3+ neutrophil count decrease based on lab values (NEU)
 c) any grade adjudicated drug-related interstitial lung disease (ILD)
 d) Adverse events leading to discontinuation (AEDC)
 e) Adverse events leading to dose reduction (AEDR)
 f) Adverse events leading to dose interruption (AEDI)

CONCLUSIONS

- ER analyses showed that the 5.6 mg/kg Q3W regimen offers a positive benefit-risk profile with clinically meaningful efficacy and overall manageable and tolerable safety profile.
 - There was a positive trend in the ER relationship between anti-HER3-ac-DXd Cavgc Cycle 1 to 3 and confirmed ORR by BICR.
 - Although the probability of platelet and neutrophil count decrease, ILD, AEDC, AEDR, and ARDI increased with increasing dose, the overall safety profile is manageable and tolerable as evidenced by the low rate of ILD and AEDC.
- The analyses results support the selection of 5.6 mg/kg Q3W as the recommended dose for further clinical development of HER3-DXd monotherapy.

INTRODUCTION

- Patritumab deruxtecan (U3-1402, HER3-DXd) is a HER3-targeting antibody-drug conjugate with a topoisomerase I inhibitor payload (DXd). The drug-to-antibody ratio is approximately 8
- HER3-DXd is currently being investigated as an anticancer agent in several phase 1-3 clinical studies in breast cancer (BC) and non-small cell lung cancer (NSCLC) patients. HER3-DXd has demonstrated significant antitumor activity in HER3-expressing EGFR-mutated, TKI-resistant, patient-derived xenograft models and has been well-tolerated in early clinical readouts
- It is currently being developed as an anticancer agent for treating multiple tumor types including NSCLC and BC

METHODS

Data

- **The efficacy analysis population consisted of 136 response evaluable NSCLC patients (Dose range: 3.2-6.4 mg/kg Q3W) from 1 phase I study**
- Study U31402-A-U102 is a multicenter, open-label, Phase 1 study of HER3-DXd in subjects with metastatic or unresectable NSCLC
 - The dose regimens were 3.2, 4.8, 5.6 and 6.4 mg/kg Q3W; up-titration of 3.2, 4.8 and 6.4 mg/kg on day 1 of first 3 cycles, followed by 6.4 mg/kg Q3W
 - The primary objectives for dose escalation part was to assess the safety and tolerability of U3-1402 in metastatic or unresectable NSCLC subjects, and to determine the recommended dose for expansion (RDE) of U3-1402 in metastatic or unresectable EGFRm NSCLC subjects who (a) are T790M mutation-negative after disease progression during treatment
 - The primary objective for dose expansion part was to investigate the antitumor activity of U3-1402
- **The safety analysis population consisted of 400 cancer patients (Dose range: 1.6- 8 mg/kg Q3W) with evaluable exposure from multiple BC and NSCLC studies (U31402-A-U102 and U31402-A-J101)**
- Study U31402-A-J101 is a Phase 1/2, multicenter, open-label, multiple dose, first-in-human study of HER3-DXd in subjects with HER3-positive metastatic BC
 - The dose regimens were 1.6, 3.2, 4.8, 6.4, and 8 mg/kg Q3W; 4.2 mg/kg Q2W in 14-day cycles for 3 cycles, followed by 6.4 mg/kg Q3W; up-titration of 3.2, 4.8 and 6.4 mg/kg on day 1 of first 3 cycles, followed by 6.4 mg/kg Q3W
 - The primary objectives for dose escalation part was to assess safety and tolerability of U3-1402, to determine the maximum tolerated dose (MTD) of U3-1402 and to determine the recommended dose(s) for expansion (RDEs) of U3-1402
 - The primary objectives for dose expansion part was to assess safety and evaluate efficacy of U3-1402 at the RDEs in subjects with human epidermal growth factor receptor 2 (HER2)-negative and hormone receptor (HR)-positive breast cancer

Table 1. Methods

Study	Title	Dose Regimens	Primary Objectives	Analysis
U31402-A-U102	multicenter, open-label, Phase 1 study of HER3-DXd in subjects with metastatic or unresectable NSCLC	3.2, 4.8, 5.6 and 6.4 mg/kg Q3W; up-titration of 3.2, 4.8 and 6.4 mg/kg on day 1 of first 3 cycles, followed by 6.4 mg/kg Q3W	• Dose escalation part: to assess the safety and tolerability of U3-1402 in metastatic or unresectable NSCLC subjects, and to determine the recommended dose for expansion (RDE) of U3-1402 in metastatic or unresectable EGFRm NSCLC subjects who (a) are T790M mutation-negative after disease progression during treatment • Dose expansion part: to investigate the antitumor activity of U3-1402	ER efficacy and safety
U31402-A-J101	Phase 1/2, multicenter, open-label, multiple dose, first-in-human study of HER3-DXd in subjects with HER3-positive metastatic BC	The dose regimens were 1.6, 3.2, 4.8, 6.4, and 8 mg/kg Q3W; 4.2 mg/kg Q2W in 14-day cycles for 3 cycles, followed by 6.4 mg/kg Q3W; up-titration of 3.2, 4.8 and 6.4 mg/kg on day 1 of first 3 cycles, followed by 6.4 mg/kg Q3W.	• Dose escalation part: to assess safety and tolerability of U3-1402, to determine the maximum tolerated dose (MTD) of U3-1402 and to determine the recommended dose(s) for expansion (RDEs) of U3-1402 • Dose expansion part: to assess safety and evaluate efficacy of U3-1402 at the RDEs in subjects with human epidermal growth factor receptor 2 (HER2)-negative and hormone receptor (HR)-positive breast cancer	ER safety

Summary of event rate in safety analysis.

AE	n	Event	
		True	False
J101			
Gr.3+ platelet count decrease	181	59 (32.6)	122 (67.4)
Gr.3+ neutrophil count decrease	181	21 (11.6)	160 (88.4)
Dose discontinuation	181	11 (6.1)	170 (93.9)
Dose interruption	181	91 (50.3)	90 (49.7)
Dose reduction	181	30 (16.6)	151 (83.4)
Interstitial lung disease	181	8 (4.4)	173 (95.6)
U102			
Gr.3+ platelet count decrease	219	40 (18.3)	179 (81.7)
Gr.3+ neutrophil count decrease	219	39 (17.8)	180 (82.2)
Dose discontinuation	219	17 (7.8)	202 (92.2)
Dose interruption	219	66 (30.1)	153 (69.9)
Dose reduction	219	46 (21.0)	173 (79.0)
Interstitial lung disease	219	12 (5.5)	207 (94.5)

Summary is count (percent).
n: number of records summarized.

Model Development

- Logistic regression models were used to describe the ER relationships for both the efficacy and safety endpoints
- Average concentration since first dose to the response or censored to the last dose/dosing interval (Cavgc) of anti-HER3-ac-DXd was used for the exposure-efficacy analysis
- For each safety endpoint, anti-HER3-ac-DXd or DXd Cavgc (till AE or censoring at 21 days after the last treatment date), and maximum concentration in the first dosing interval (Cmax1) were tested. The exposure metric that best described the ER relationship was selected using the LOO Information Criterion (LOOIC)
- For each analysis, base model development evaluated log-linear and sigmoidal Emax structural models. The full model included all the pre-specified covariates and their main effects on the log odds. Covariate interaction effects with exposure was not examined
- The final model was obtained by including only the covariate effects that were considered statistically significant by backwards elimination from the full model.
- Investigated baseline covariates:
 - **Efficacy:** age, weight, tumor size, sex, ECOG, race, bone metastasis, liver metastasis, brain metastasis, membrane H score, and the use of prior chemotherapy
 - **Safety:** age, weight, sex, platelet count decrease, neutrophil count decrease, hemoglobin (for neutrophil count decrease and platelet count decrease), ECOG, race, liver metastasis, membrane H score, use of prior IO therapy and tumor type

Model Evaluations

- Logistic model evaluations were performed using posterior predictive checks (PPC)
- The proportion of responders (for ER efficacy) or subjects with AEs (for ER safety) were simulated across the posterior samples of model parameters. The simulated posterior predictive distributions were summarized by exposure quartile and key dose groups and compared to observed proportions

Model-based Simulation

- Virtual patient populations with 200 patients were created by randomly sampling with replacement from the NSCLC patients
- Exposures of anti-HER3-ac-DXd and DXd were simulated at the 4.8, 5.6, 6.4 mg/kg Q3W and the up-titration dosing regimens
- The responses were simulated and summarized across joint posterior distribution of ER model parameters

RESULTS

ER Efficacy

- The objective response rate increased with increasing anti-HER3-ac-DXd exposure (**Figure 1**)
- There was a positive trend in the ER relationship between anti-HER3-ac-DXd Cavgc and confirmed ORR by BICR (**Figure 1**)
- Among the exploratory variables considered, the only predictive covariate was prior chemotherapy (patients with prior chemotherapy resulted in a 28.4% decrease in odds of being a responder compared to patients without prior chemotherapy) (**Figure 2**)
- Simulations of ORR across varying treatment regimens (4.8 mg/kg Q3W, 5.6 mg/kg Q3W, and 6.4 mg/kg Q3W) demonstrated a positive dose-response relationship, while up-titration regimen showed lower ORR efficacy (**Figure 2**)

Figure 1: Predicted Probability of Overall Response as a Function of Anti-HER3-ac-DXd Cavgc[a,b]

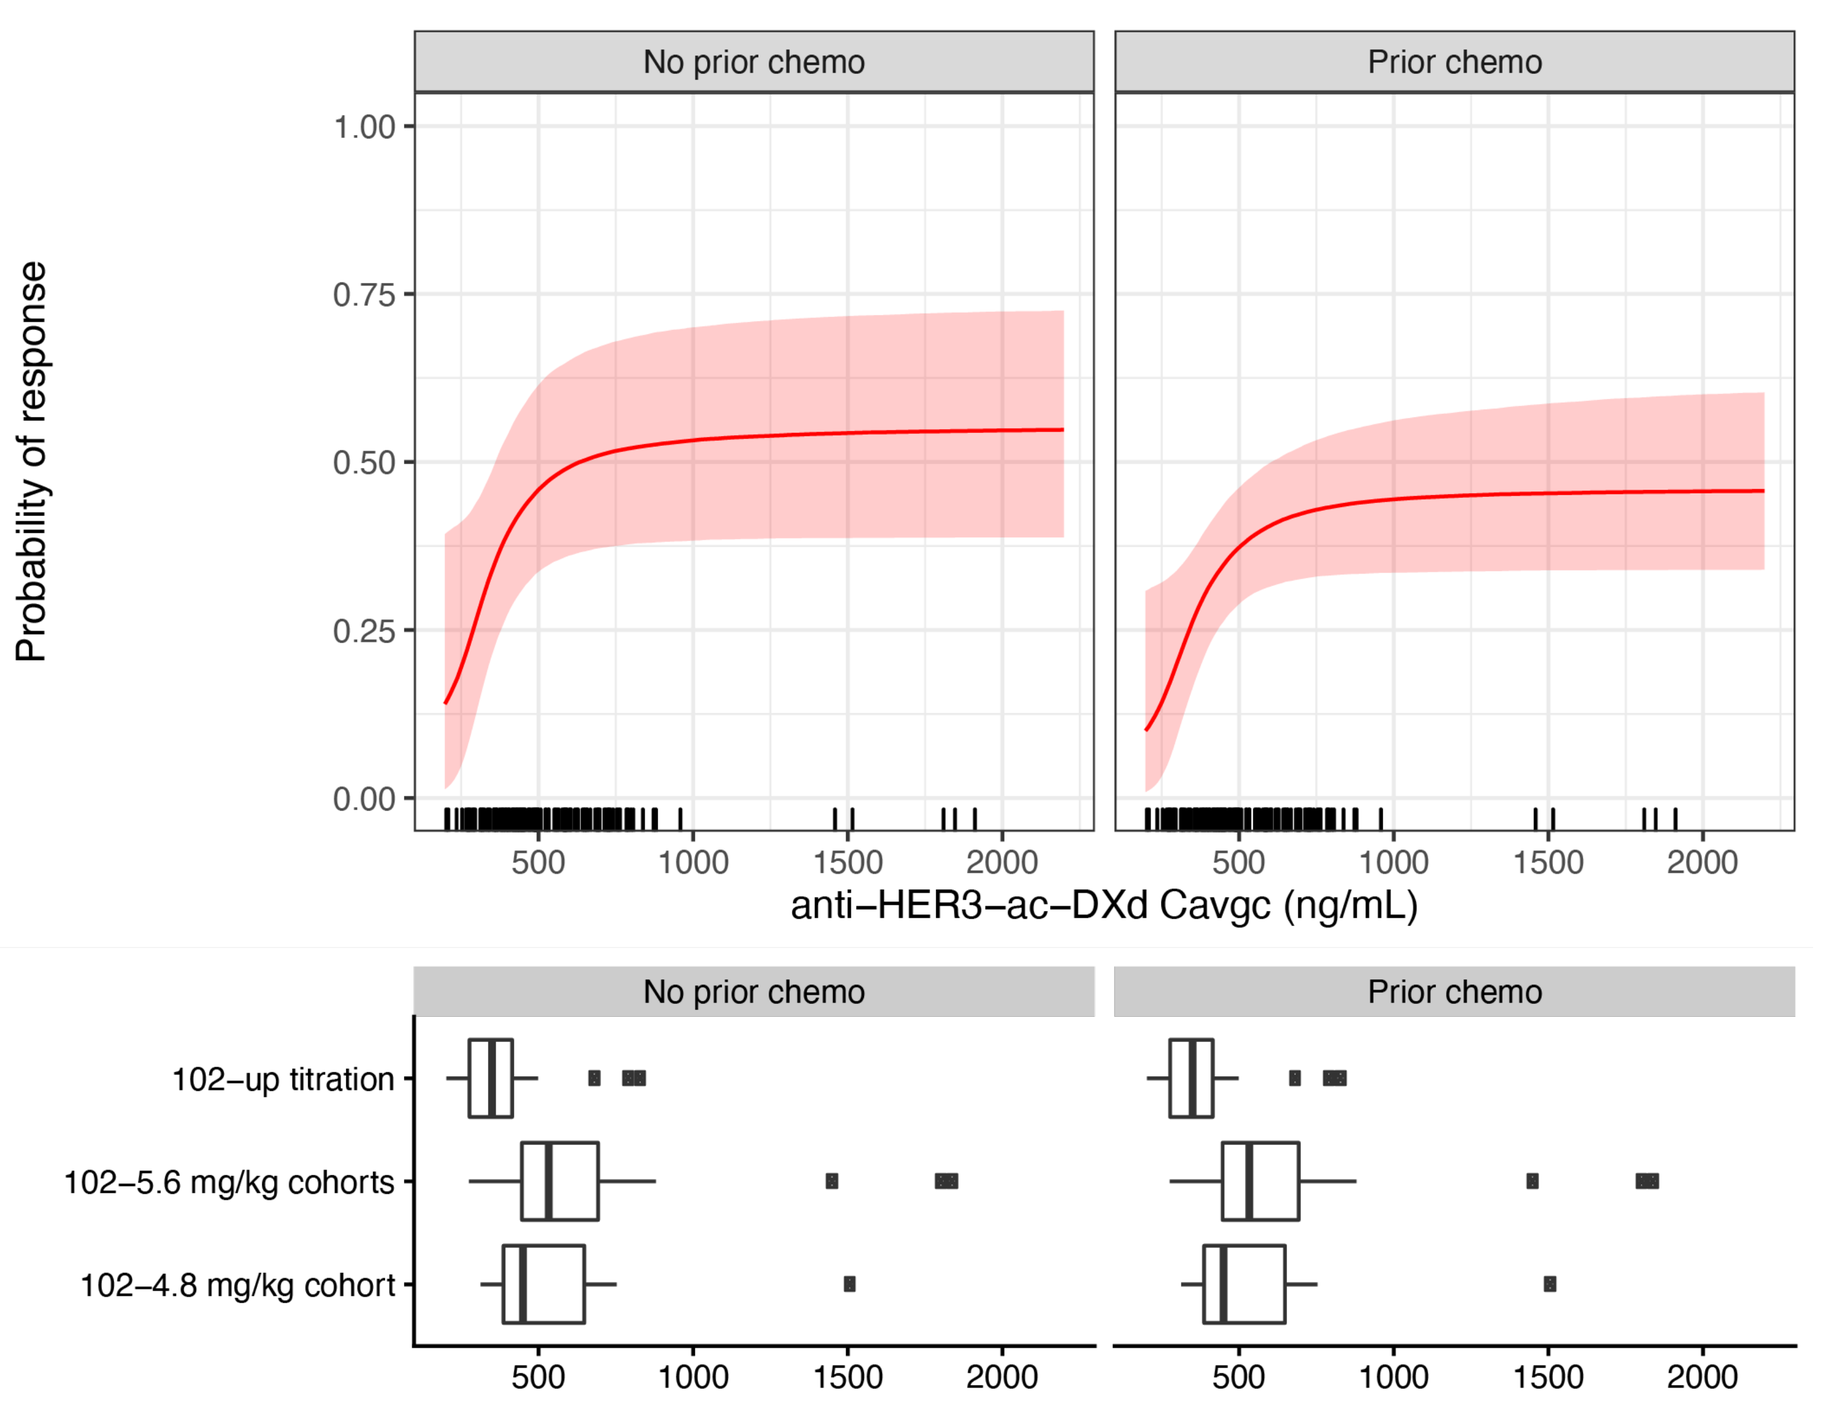
No prior chemo
Prior chemo
Probability of response
anti−HER3−ac−DXd Cavgc (ng/mL)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohort

Figure 2: Posterior Predictive Distribution for Proportion of Partial or Complete Responders in Each Quartile of Anti-HER3-ac-DXd Cavgc. The Vertical Dashed Red Lines Represent Observed Proportions

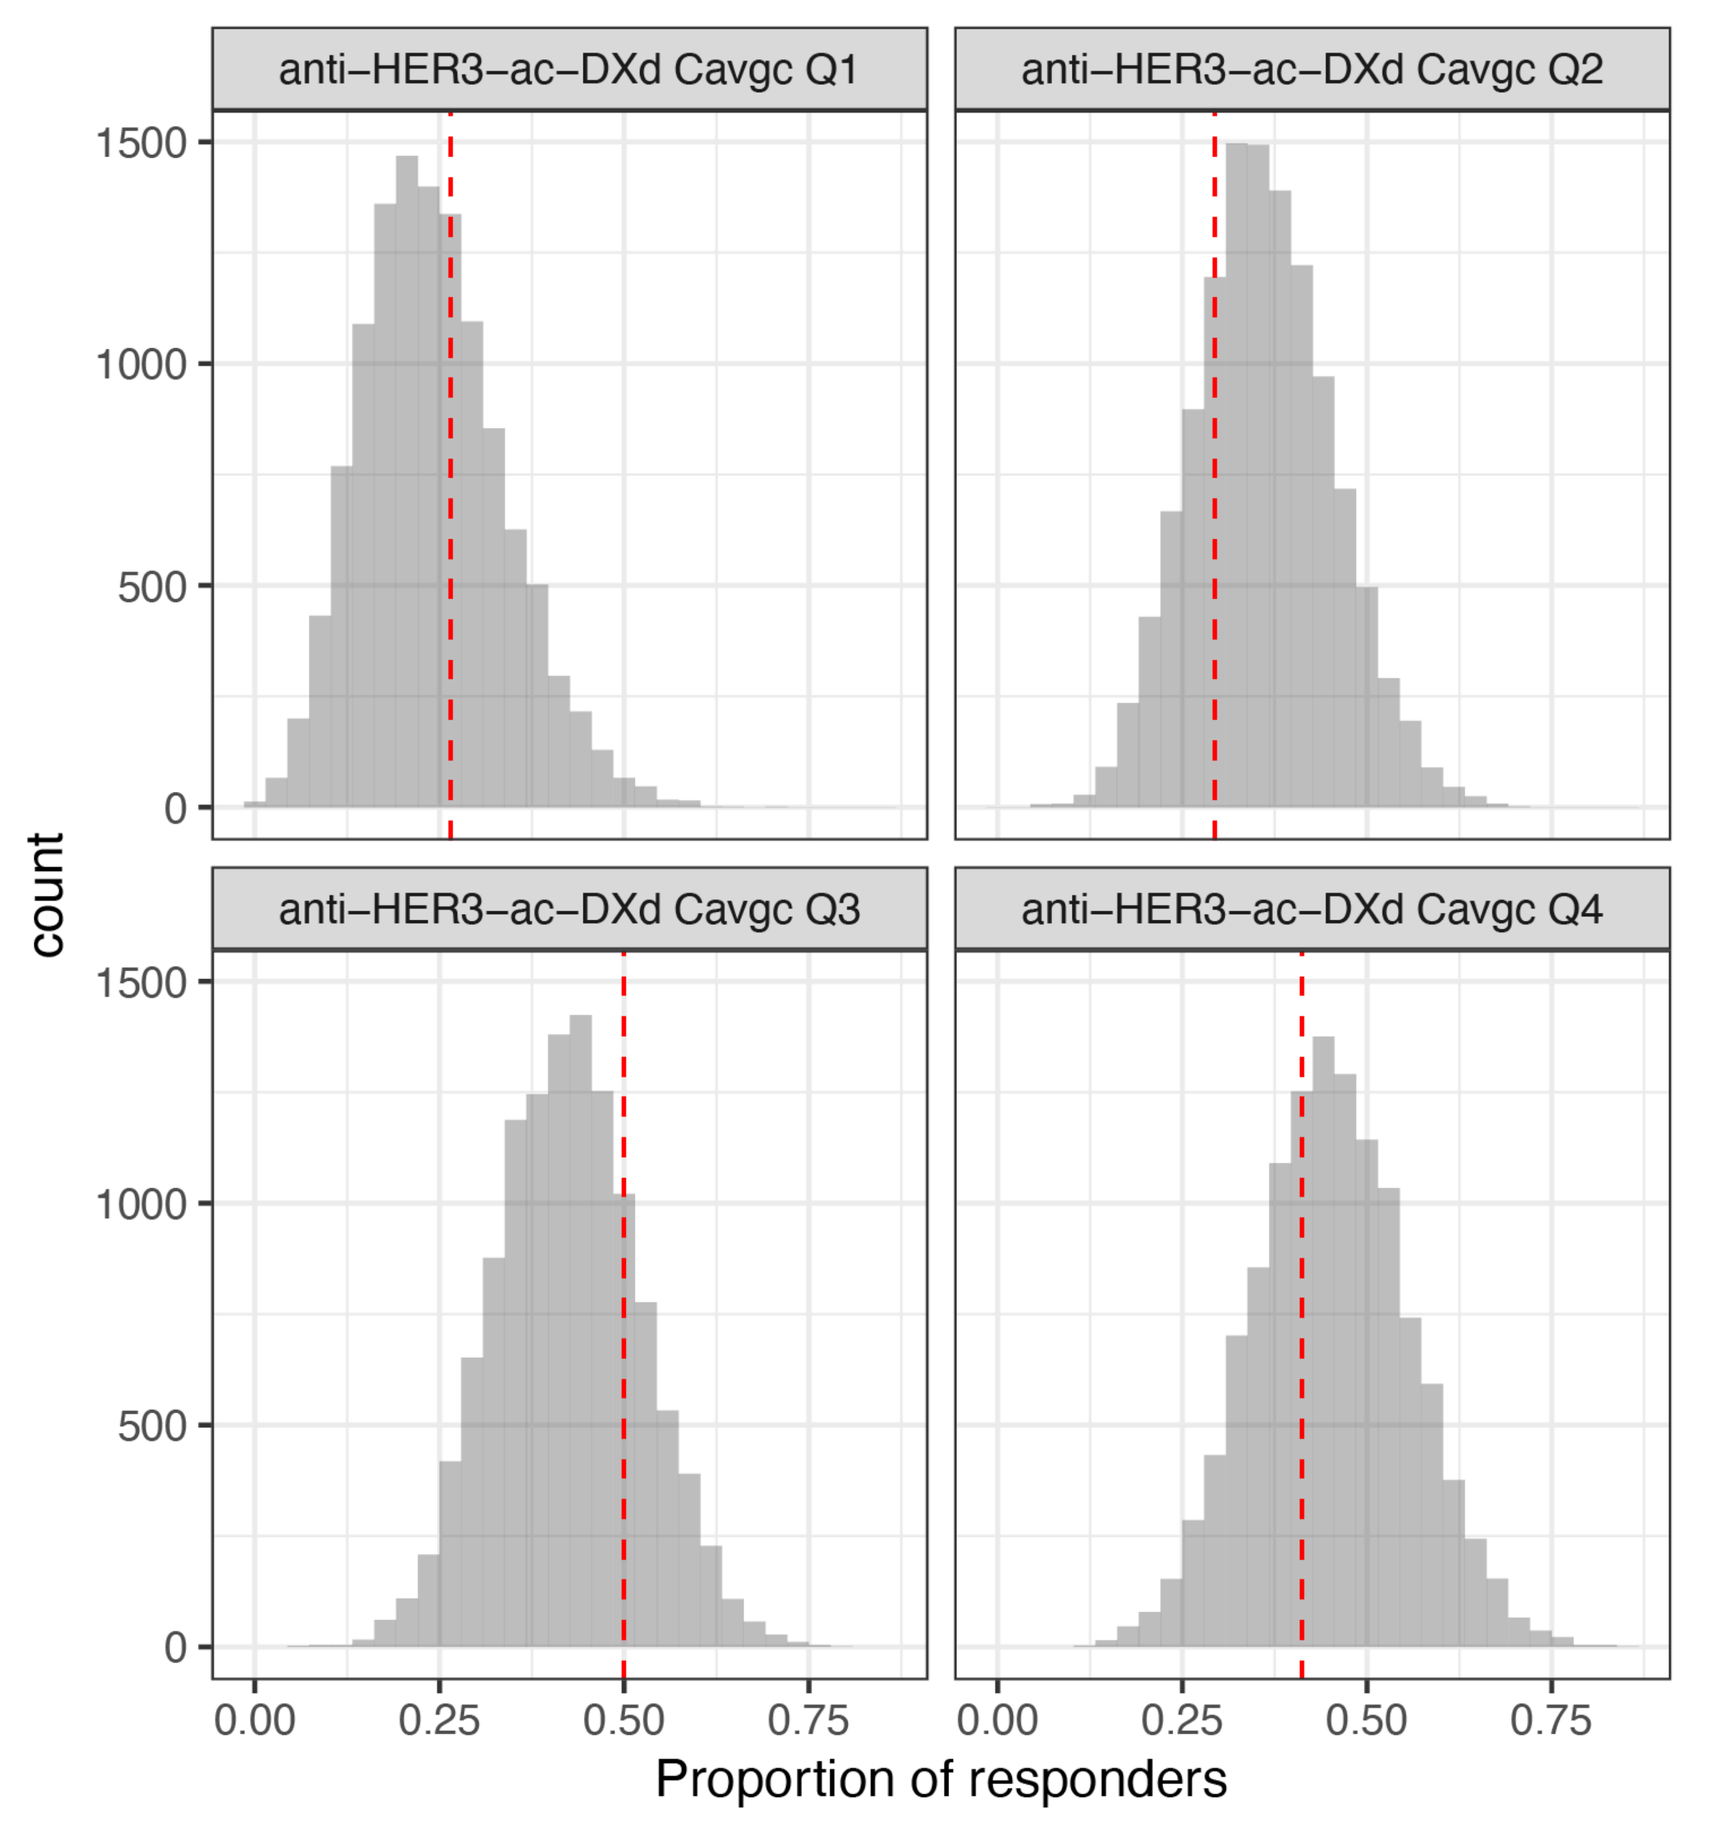
anti−HER3−ac−DXd Cavgc Q1
anti−HER3−ac−DXd Cavgc Q2
anti−HER3−ac−DXd Cavgc Q3
anti−HER3−ac−DXd Cavgc Q4
count
Proportion of responders

The model adequately captured the ER relationship of ORR, as demonstrated by the simulated distribution of responders encompassing the observed median in each quartile of anti-HER3-ac-DXd Cavgc.

ER Safety

Table 2. Predictive Exposure metrics and covariates for safety endpoints

AE Endpoint	Predictive Exposure	Predictive Covariates
Gr.3+ platelet count decrease	DXd Cmax1 (log-linear)	race (Asian vs. non-Asian) and baseline platelet count
Gr.3+ neutrophil count decrease	DXd Cmax1 (log-linear)	age
Drug-related ILD	Anti-HER3-ac-DXd Cavg (sigmoidal Emax)	none
AEDC	DXd Cavg (log-linear)	none
AEDR	DXd Cavg (log-linear)	tumor type (BC vs. NSCLC)
AEDI	DXd Cavg (log-linear)	age, race and ECOG (>=1 vs. 0)

- Increasing DXd Cmax1 was positively associated with higher risk of Gr.3+ platelet count decrease (**Figure 3**)
 - The probability was higher in Asian patients or patients with lower baseline platelet count
- Increasing DXd Cmax1 was positively associated with higher risk of Gr.3+ neutrophil count decrease (**Figure 4**)
 - The probability increased with increasing age

Figure 3: Predicted Probability of Gr.3+ platelet count decrease as a Function of Cycle 1 DXd Cmax (Top panel: by Asian vs Non-Asian Race, Bottom Panel: Baseline Platelet Count[a,b]

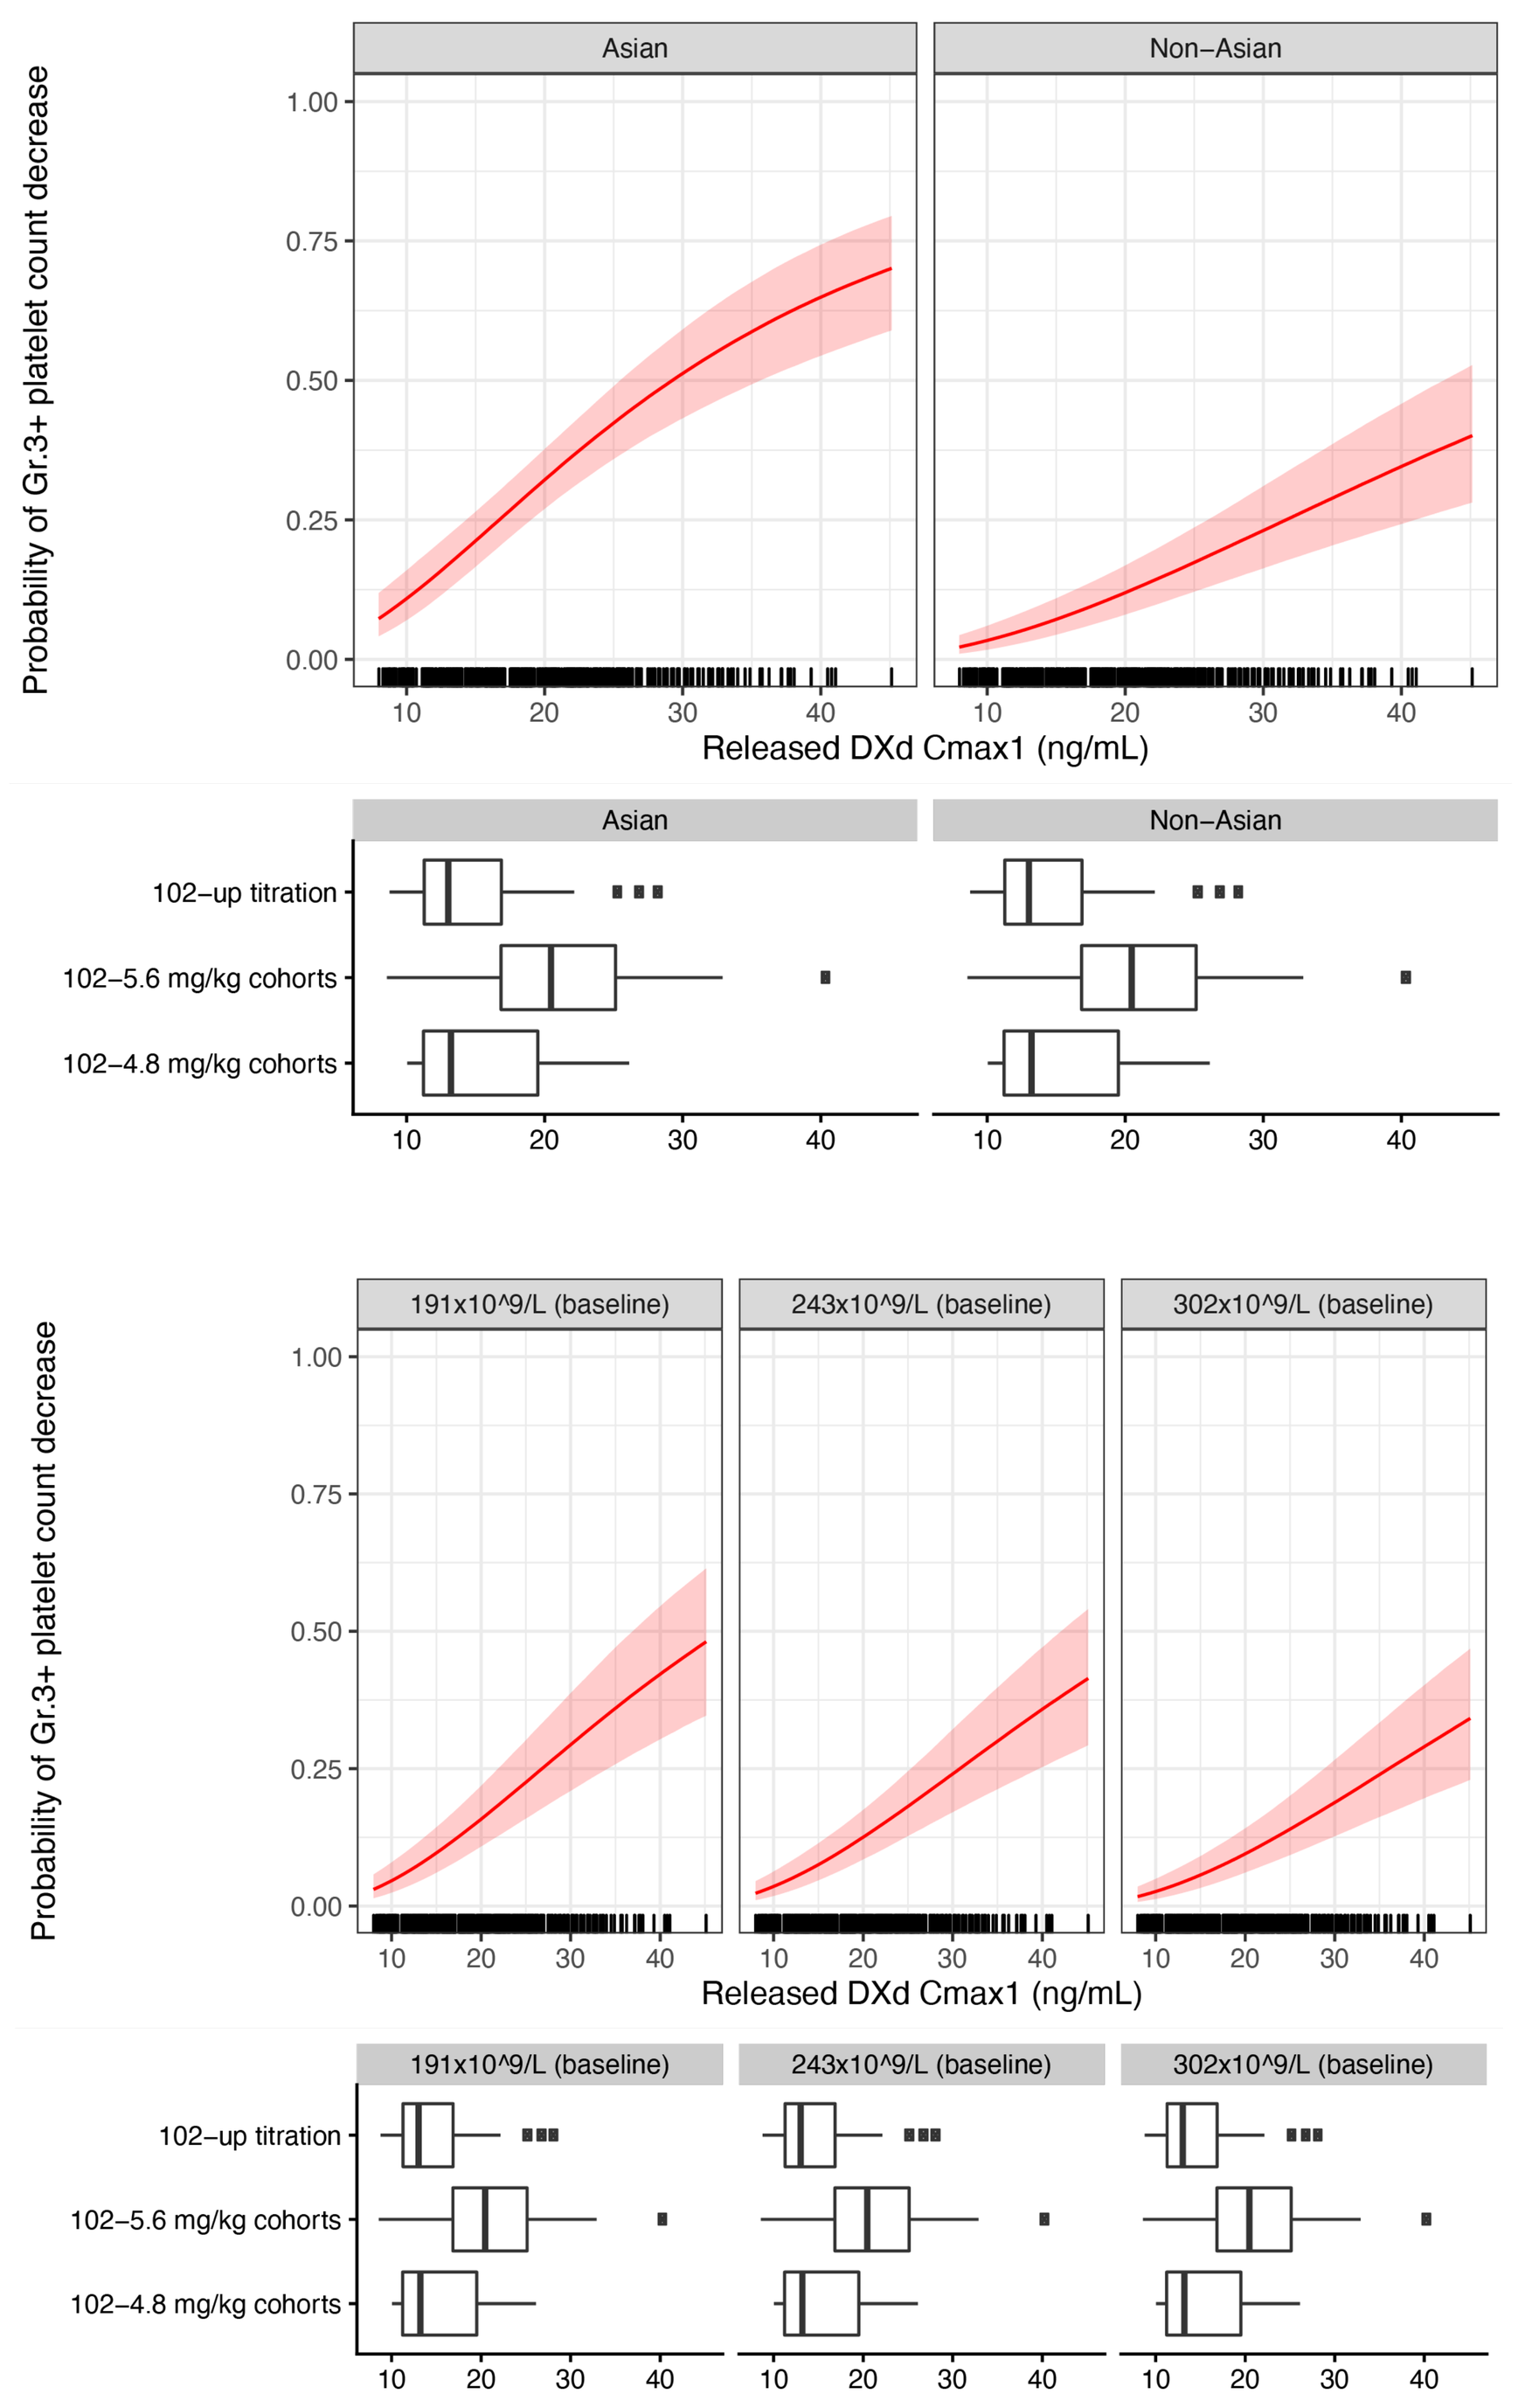
Asian
Non−Asian
Probability of Gr.3+ platelet count decrease
Released DXd Cmax1 (ng/mL)
191x10^9/L (baseline)
243x10^9/L (baseline)
302x10^9/L (baseline)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohorts

The presented baseline platelet count values represent the 25th, median and 75th percentiles in the analysis dataset.
The probability of Gr.3+ TCP increased with increasing DXd Cmax in Cycle1.
The probability was higher in Asian patients or patients with higher baseline platelet counts.

Figure 4: Predicted Probability of Gr.3+ neutrophil count decrease as a Function of Cycle 1 DXd Cmax[b]

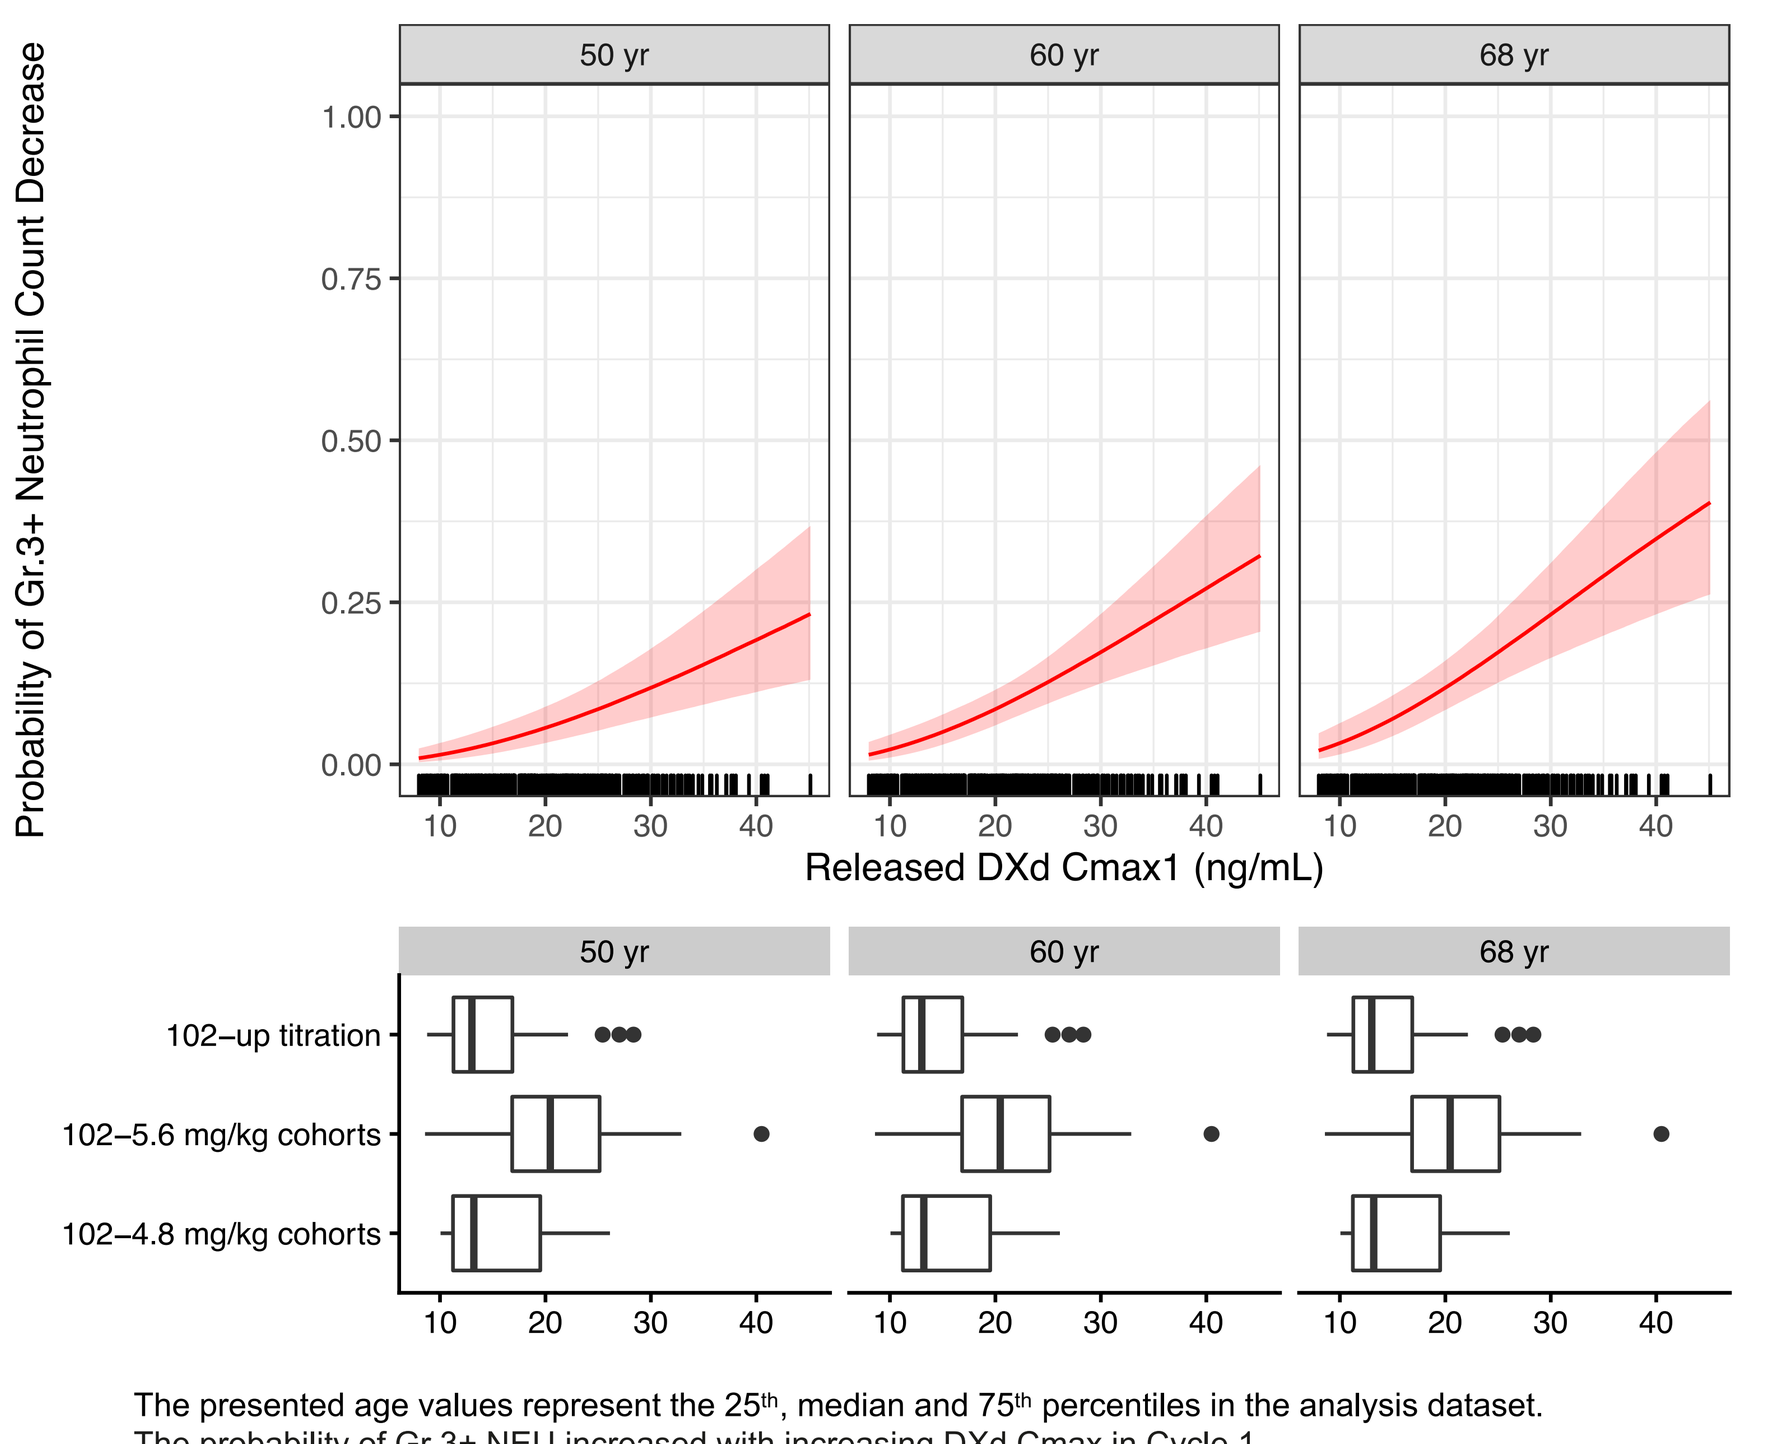
50 yr
60 yr
68 yr
Probability of Gr.3+ Neutrophil Count Decrease
Released DXd Cmax1 (ng/mL)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohorts

The presented age values represent the 25th, median and 75th percentiles in the analysis dataset.
The probability of Gr.3+ NEU increased with increasing DXd Cmax in Cycle 1.
The probability also increased with increasing age.

- Increasing anti-HER3-ac-DXd Cavg was positively associated with higher risk of drug-related adjudicated ILD (**Figure 5**)
 - Among the exploratory variables considered, no covariates were identified as predictive

Figure 5: Predicted Probability of Drug-Related ILD as a Function of anti-HER3-ac-DXd Average Concentration[a,b]

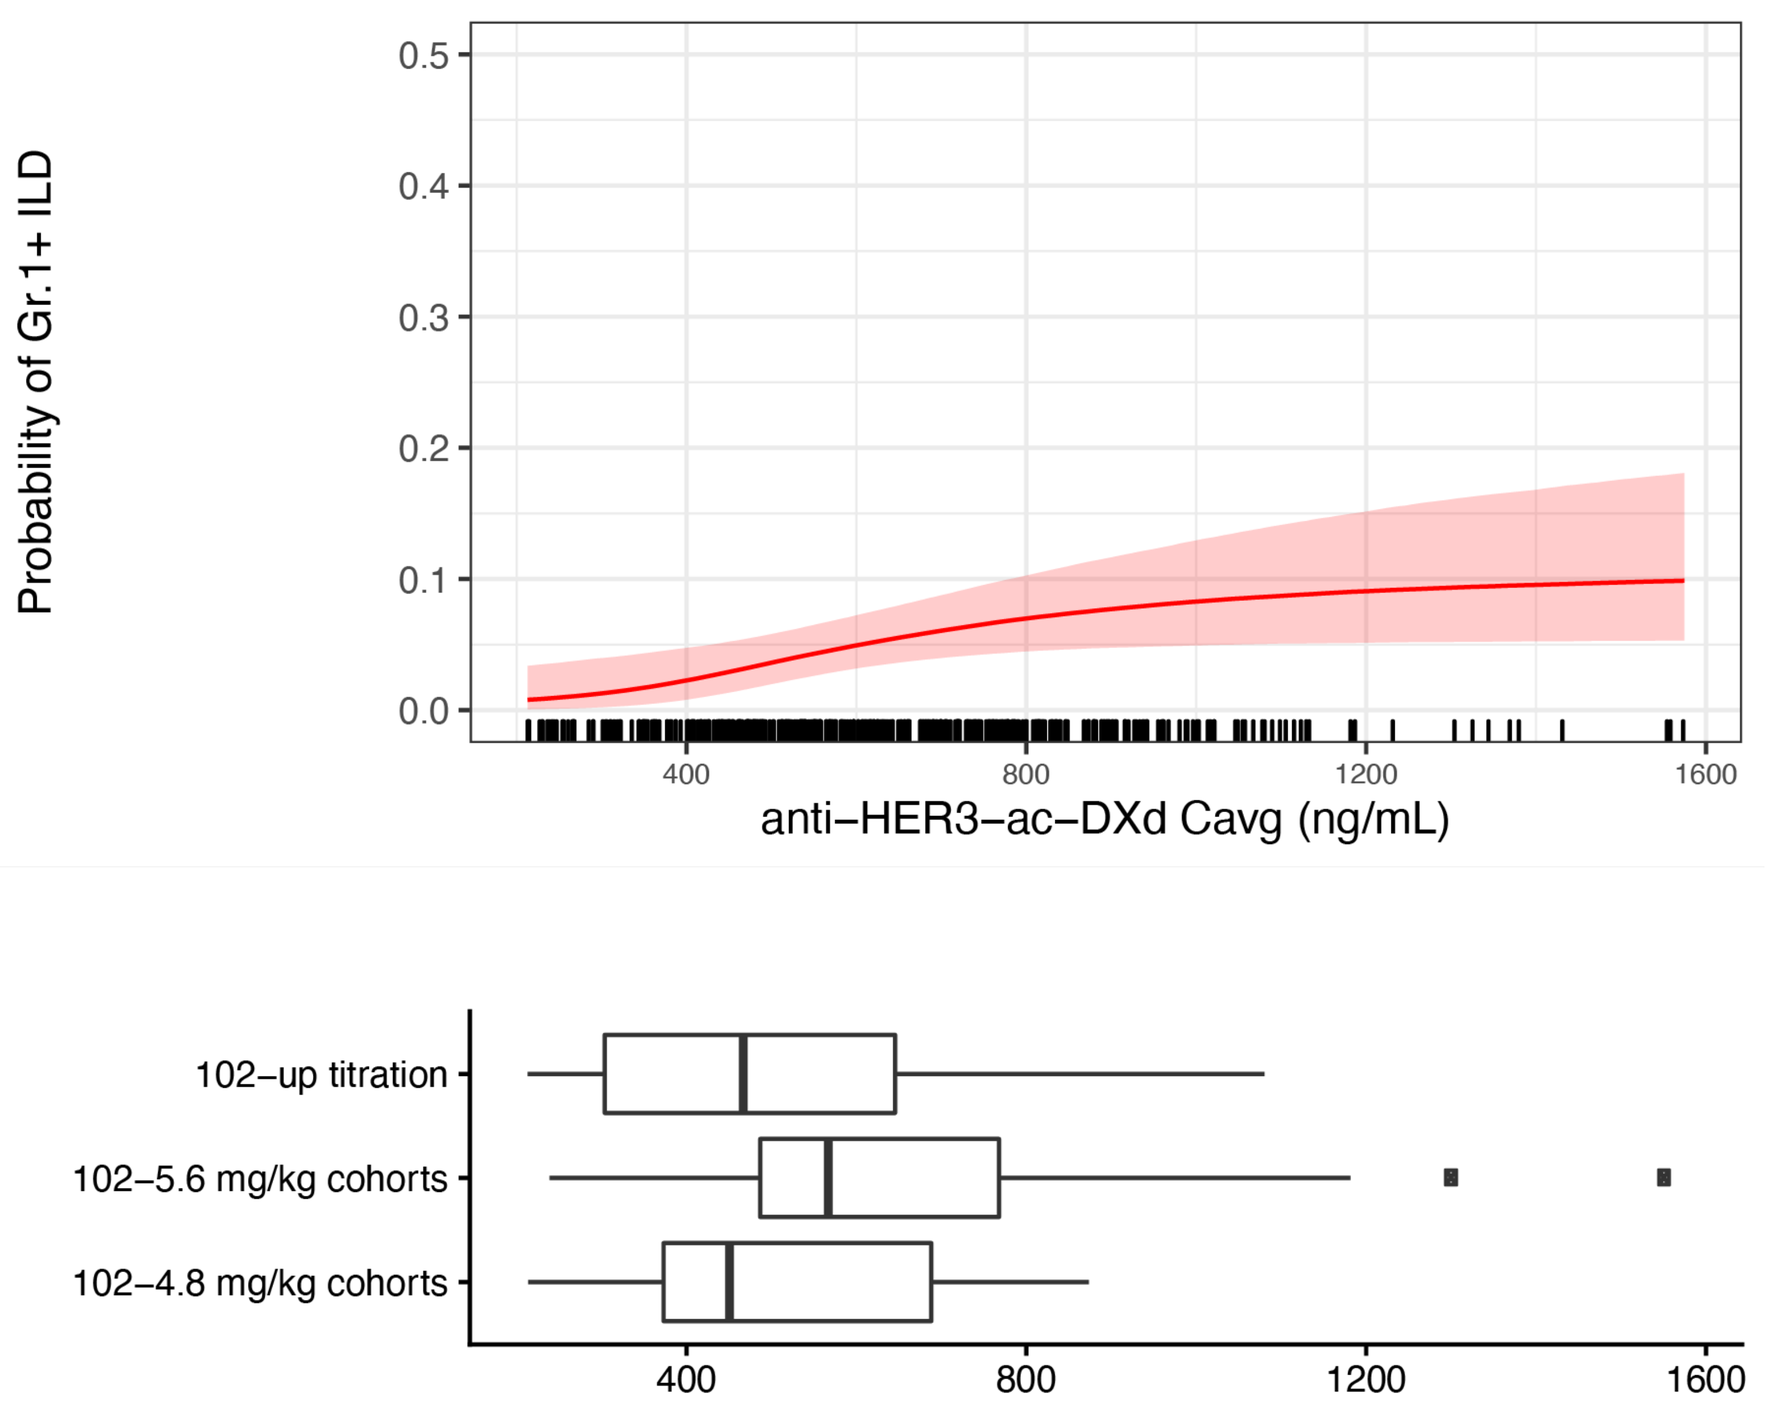
Probability of Gr.1+ ILD
anti−HER3−ac−DXd Cavg (ng/mL)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohorts

The probability of drug related ILD increased with increasing Cavg of anti-HER3-ac-DXd while probabilities of AEDC, AEDR and AEDI increased with increasing Cavg of DXd

Figure 6: Predicted Probability of Adverse Events Leading to Discontinuation as a Function of DXd Average Concentration[a,b]

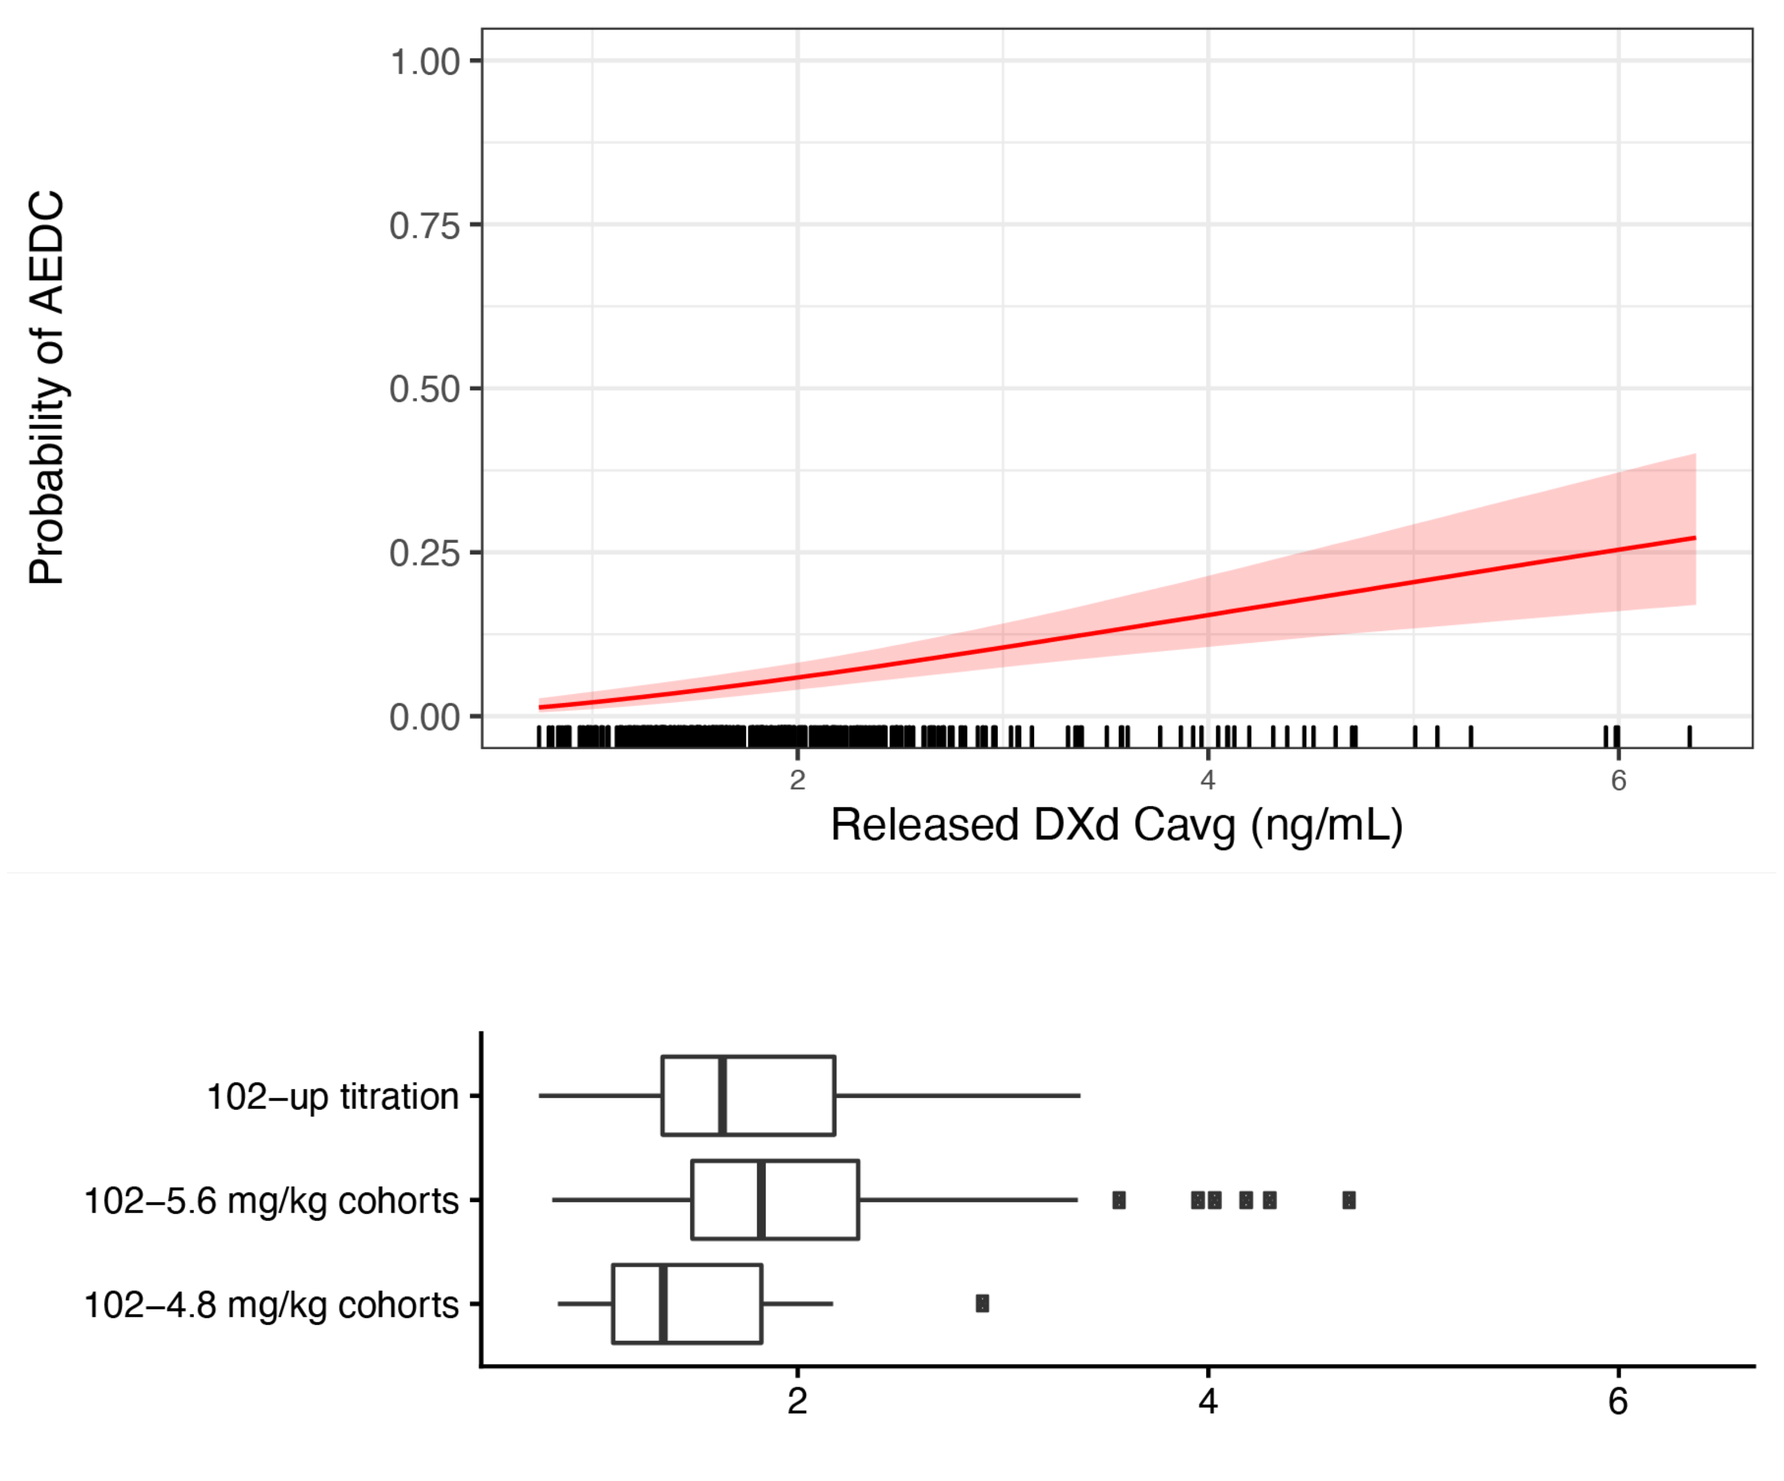
Probability of AEDC
Released DXd Cavg (ng/mL)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohorts

Figure 7: Predicted Probability of Adverse Events Leading to Dose Reduction as a Function of DXd Average Concentration by Tumor Type[a,b]

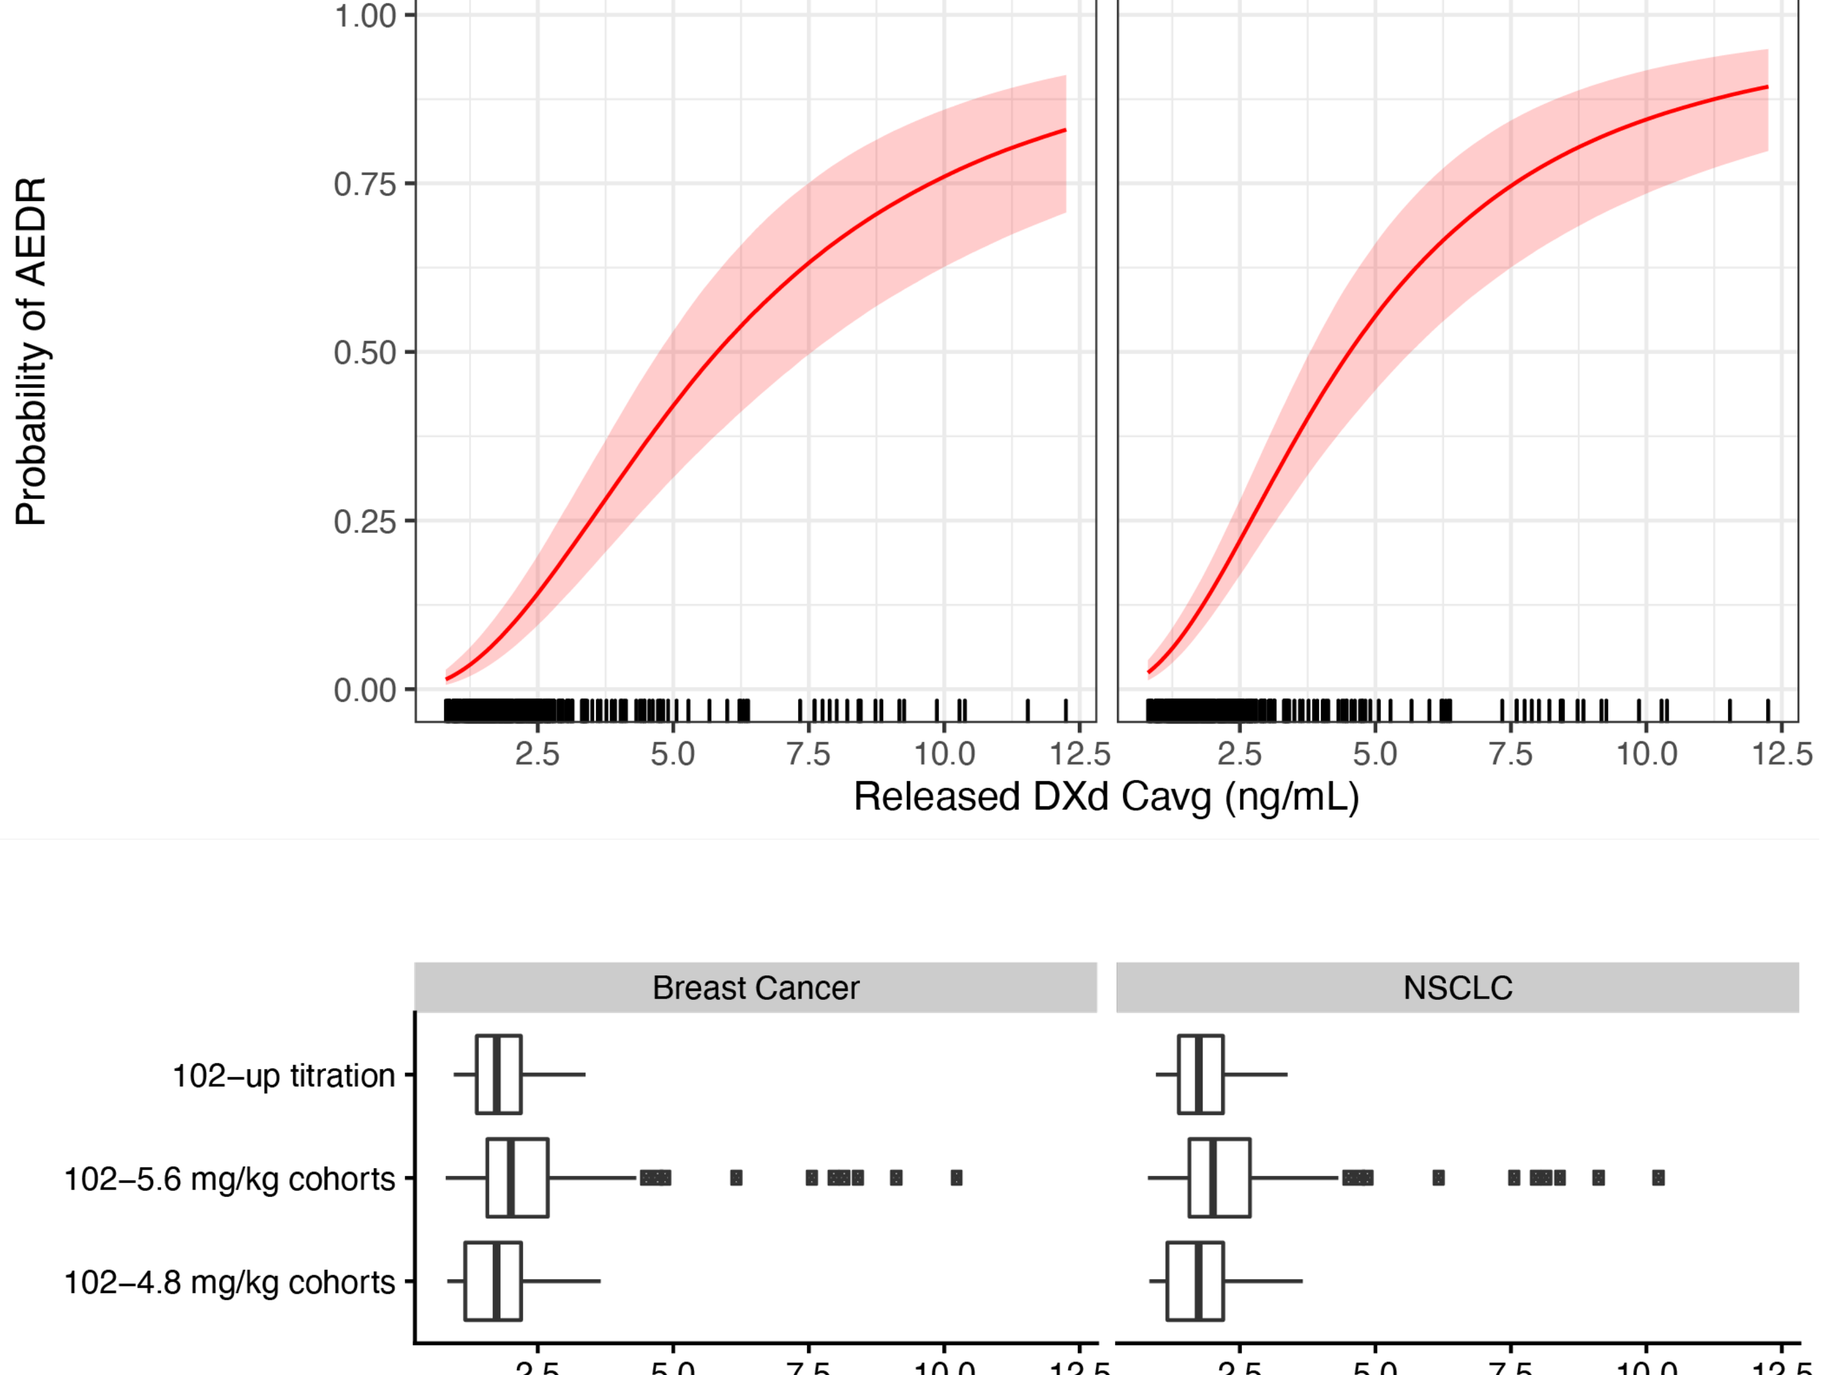
Breast Cancer
NSCLC
Probability of AEDR
Released DXd Cavg (ng/mL)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohorts

- Increasing DXd Cavg was positively associated with higher risk of AEDC (**Figure 8**)
 - Among the exploratory variables considered, no covariates were identified as predictive
- Increasing DXd Cavg was positively associated with higher risk of AEDR (**Figure 8**)
 - The probability was higher in NSCLC patients compared to BC patients
- Increasing DXd Cavg was positively associated with higher risk of AEDI (**Figure 8**)
 - The probability was higher in older patients, Asian patients or patients with ECOG 0

Figure 8: Predicted Probability of Adverse Events Leading to Dose Interruption as a Function of DXd Average Concentration by ECOG (top panel), Race(middle panel) and Age (bottom panel)[a,b]

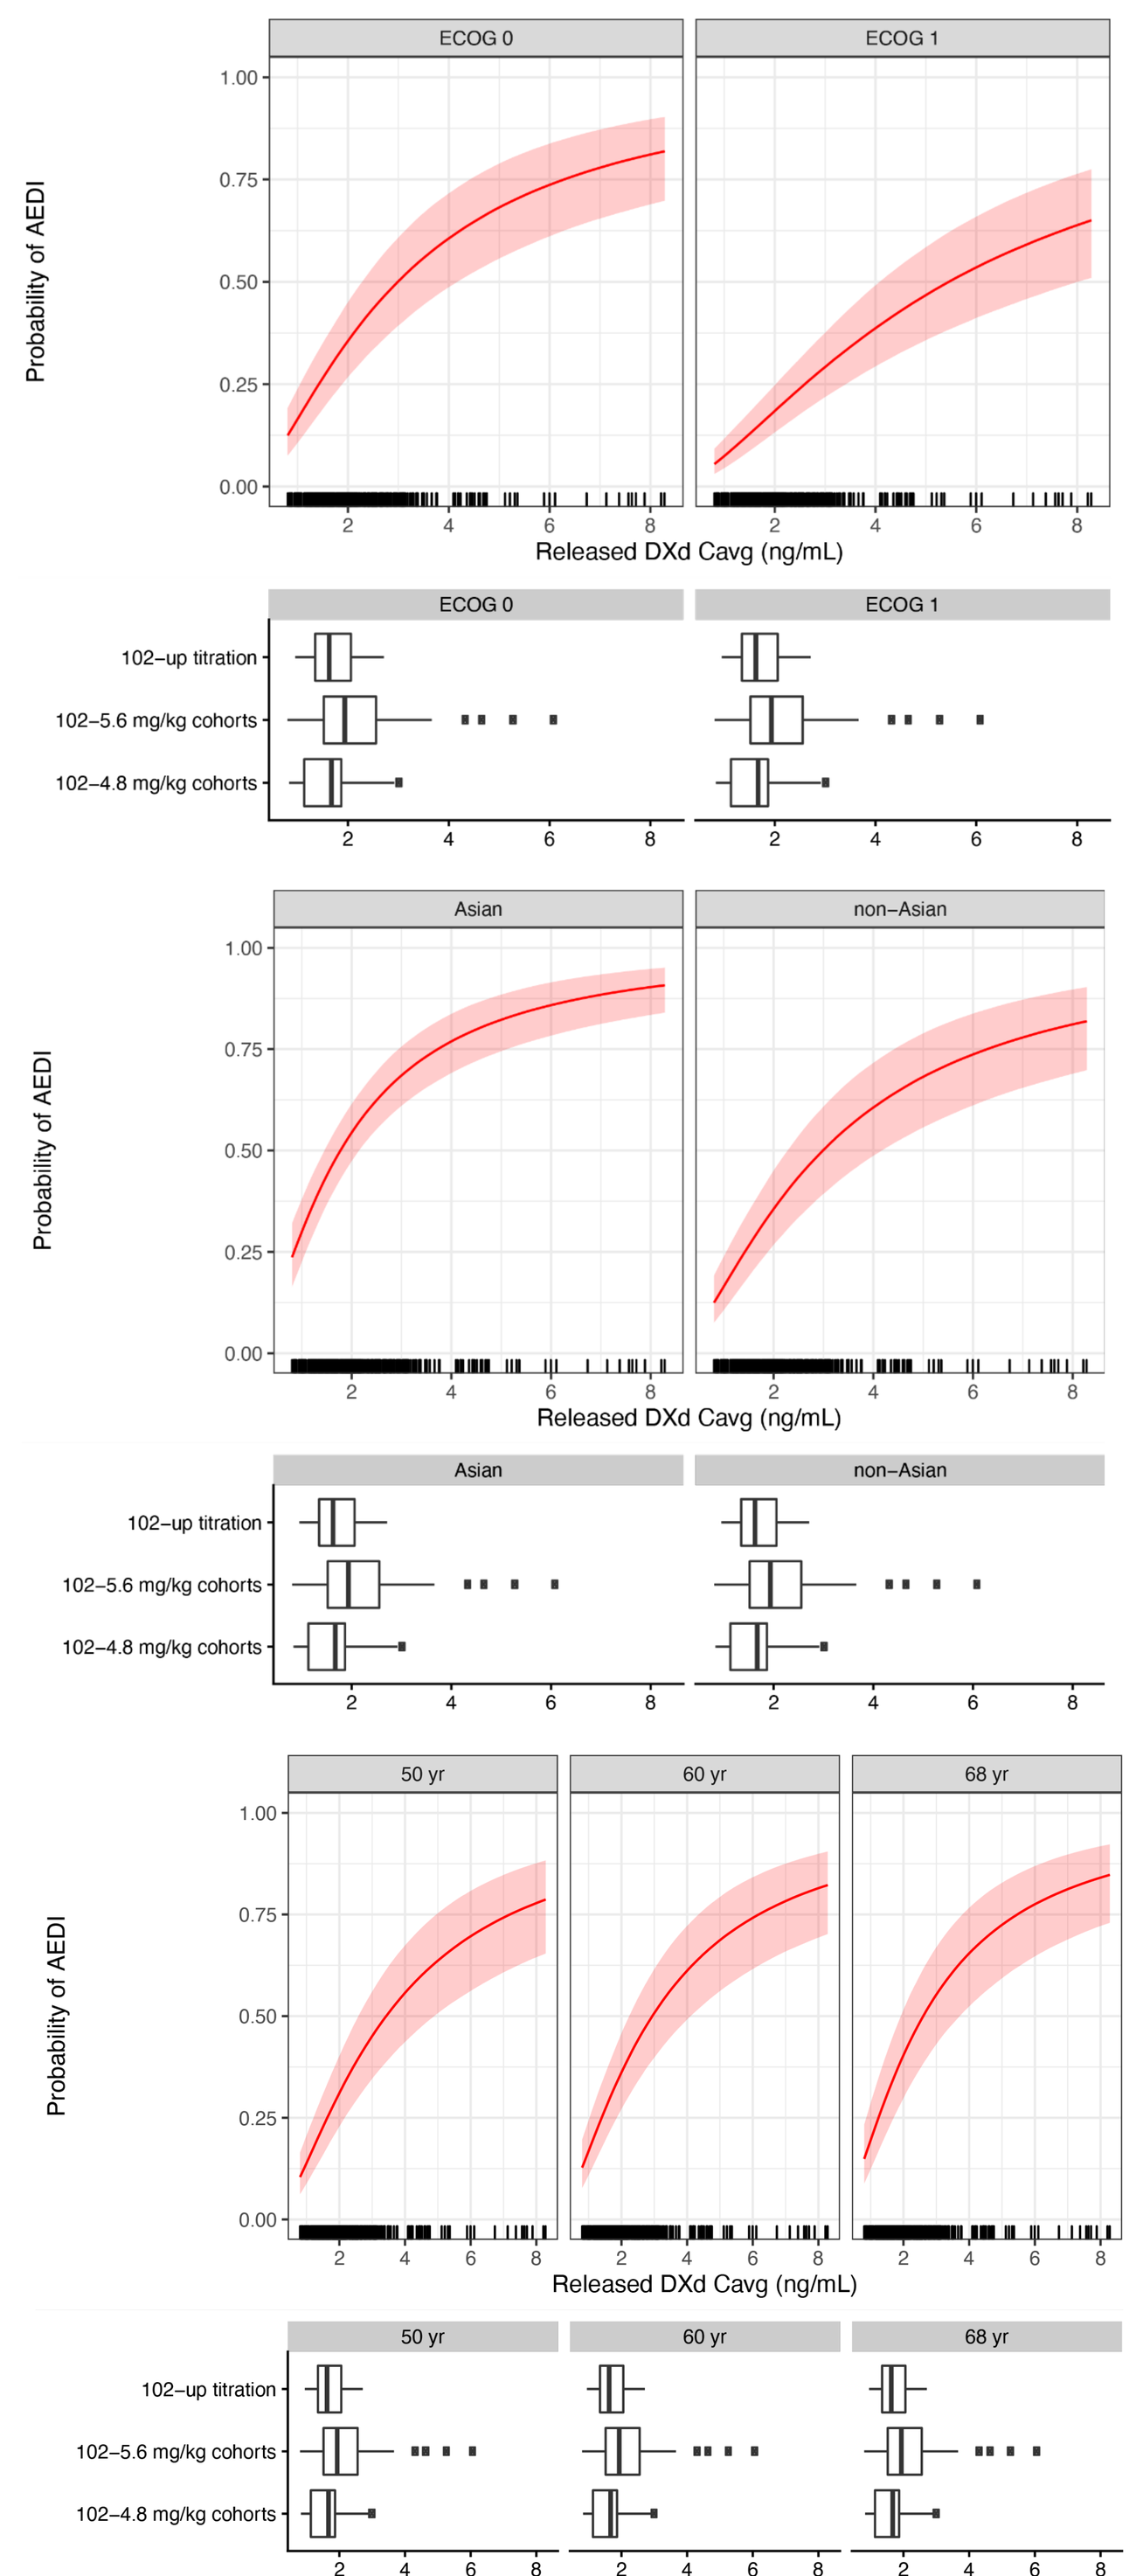
ECOG 0
ECOG 1
Asian
non−Asian
50 yr
60 yr
68 yr
Probability of AEDI
Released DXd Cavg (ng/mL)
102−up titration
102−5.6 mg/kg cohorts
102−4.8 mg/kg cohorts

Figure 9: Predicted ORR and Incidence of Safety Endpoints for HER3-DXd doses 6 mg/kg Q3W and Up-titration Regimens Using ER Analyses

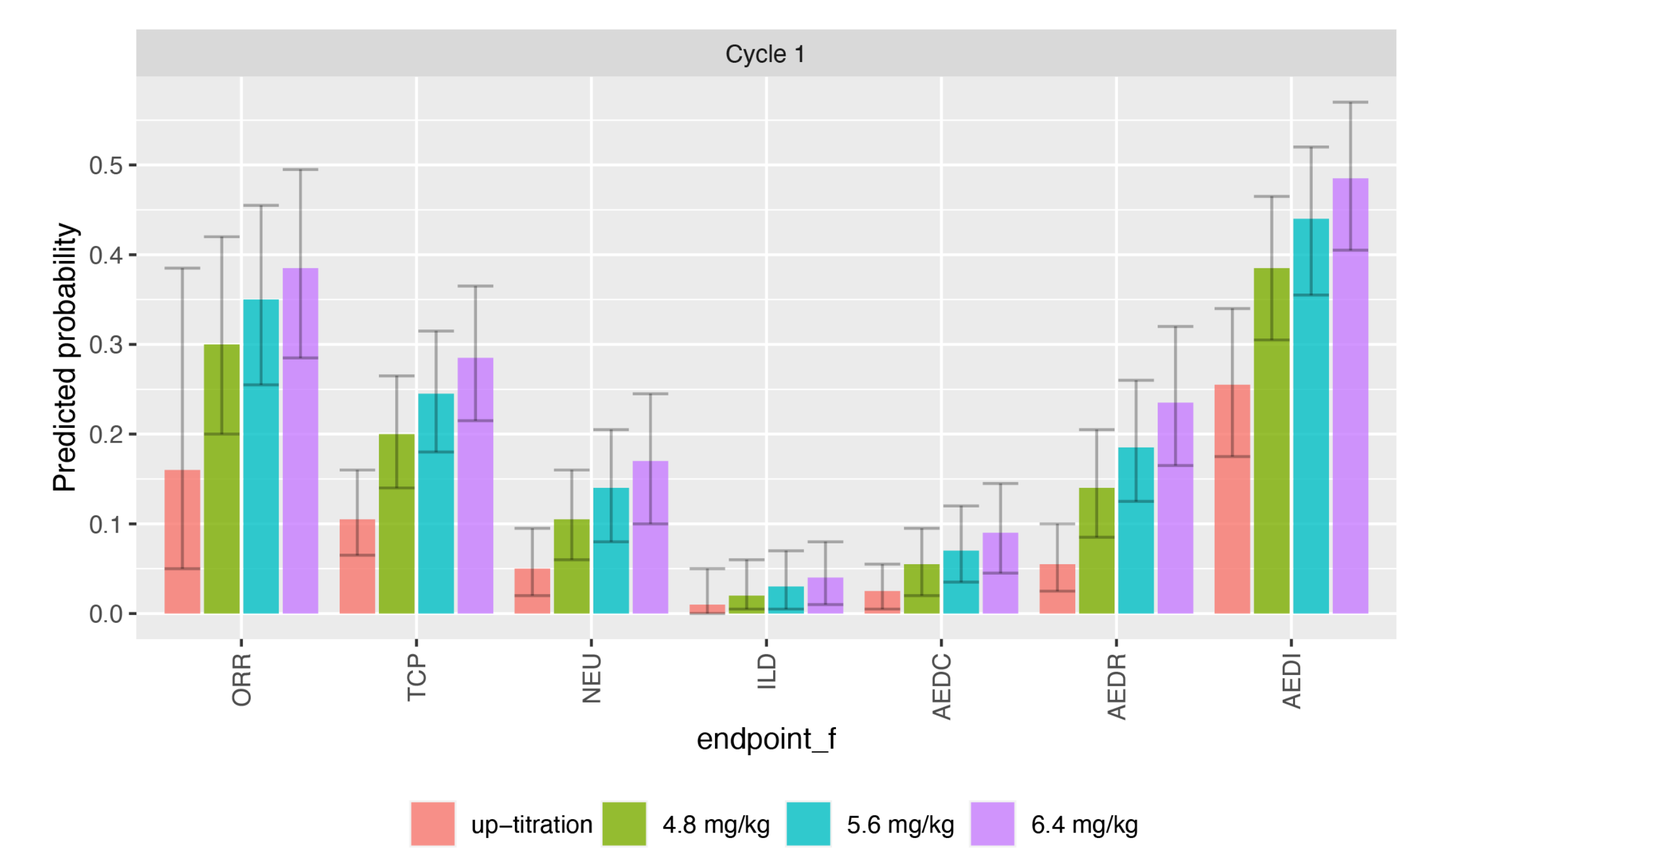
Cycle 1
Predicted probability
ORR
TCP
NEU
ILD
AEDC
AEDR
AEDI
endpoint_f
up−titration
4.8 mg/kg
5.6 mg/kg
6.4 mg/kg

5.6 mg/kg Q3W dose regimen is predicted to provide a clinically meaningfully higher ORR compared to the other dosing regimen with durable responses and manageable safety events.

Acknowledgements

We thank the patients, their families, and their caregivers for their participation. We also thank Pavan Vaddady for his valuable scientific contributions to this analysis.

Footnotes

[a] Red line is posterior median. Colored ribbon is 90% CrI. Vertical bars indicate observed individual exposure values in the overall analysis dataset. Horizontal box plot shows the distribution of the exposure values in the 3 dose regimens observed in U31402-A-U102.

[b] The sample size was 14, 84 and 29 for 4.8 mg/kg, 5.6 mg/kg and up titration cohort, respectively.

Presented at: ACoP 14, November 5-8, 2023; National Harbor, MD.